ASCO TAPUR
Targeted Agent and Profiling Utilization Registry Study

Abstract 3142: Pertuzumab plus trastuzumab in patients with solid tumors with *ERBB2* or *ERBB3* amplification or overexpression: Results from the Targeted Agent and Profiling Utilization Registry (TAPUR) Study

ASCO AMERICAN SOCIETY OF CLINICAL ONCOLOGY

EG Dib[1], M Rothe[2], PK Mangat[2], E Garrett-Mayer[2], ER Ahn[3], AS Alva[4], N Hafez[5], E Hobbs[6], M Akce[7], D Behl[8], CJ Calfa[9], TL Cannon[10], M McKean[11], F Meric-Bernstam[12], R Thota[13], A Watson[14], KJ Yost[15], DC Hinshaw[2], A Gregory[2], GN Grantham[2], S Halabi[16], RL Schilsky[2]

[1]Michigan Cancer Research Consortium, Ypsilanti, MI; [2]American Society of Clinical Oncology, Alexandria, VA; [3]City of Hope Chicago, Zion, IL; [4]University of Michigan Rogel Comprehensive Cancer Center, Ann Arbor, MI; [5]The Angeles Clinic and Research Institute, A Cedars-Sinai Affiliate, Los Angeles, CA; [6]Providence Cancer Institute, Portland, OR; [7]O'Neal Comprehensive Cancer Center, The University of Alabama at Birmingham Heersink School of Medicine, Birmingham, AL; [8]Sutter Sacramento Medical Center, Sacramento, CA; [9]UM/Sylvester Comprehensive Cancer Center, Miami, FL; [10]Inova Schar Cancer Institute, Fairfax, VA; [11]Sarah Cannon Research Institute TO, Nashville, TN; [12]University of Texas MD Anderson Cancer Center, Houston, TX; [13]Intermountain Healthcare, Murray, UT; [14]Sanford Health, Sioux Falls, SD; [15]Cancer Research Consortium of West Michigan, Grand, Rapids, MI; [16]Duke University Medical Center, Durham, NC

Background:

- TAPUR is a phase II basket study that evaluates antitumor activity of commercially available targeted agents in patients (pts) with advanced cancers with specific genomic alterations.
- **Results of a cohort of pts with various solid tumors with *ERBB2 or ERBB3* amplification (amp) or overexpression (oe) treated with pertuzumab plus trastuzumab (P+T) are reported.**

Methods:

Study Design:

- **Eligible pts:** Advanced solid tumors, ECOG performance status (PS) 0-2, adequate organ function, measurable disease, and no standard treatment (tx) options. Tx was assigned according to prespecified matching rules based on genomic tests performed in CLIA-certified, CAP-accredited laboratories selected by sites. Amp cut-offs were defined per test providers.
- Pts received P at an initial dose of 840 mg intravenously (IV) over 60 minutes (m), then 420 mg IV over 30-60 m every 3 weeks (wks) and T at an initial dose of 8 mg/kg IV over 90 m, then 6 mg/kg IV over 30-60 m every 3 wks until disease progression, unacceptable toxicity or pt or physician choice to discontinue.
- **Primary endpoint:** Disease control (DC) defined by investigator assessment of objective response (OR) or stable disease (SD) of at least 16+ wks duration (SD16+) per RECIST v1.1. Radiographic confirmation of response was not required.
- **Secondary endpoints:** OR, progression-free survival (PFS), overall survival (OS), duration of response (DOR), duration of SD, Grade 3-5 adverse events (AEs) or serious adverse events (SAEs) per CTCAE v4.0 at least possibly related to tx are reported. DOR is defined as time from pt's first documented OR to progressive disease (PD). Duration of SD is defined as time from tx start to PD.

Statistical Methods:

- Inferences are based on a one-sided exact binomial test with a null hypothesis of DC rate ≤15%; power and alpha were 86% and 0.10, respectively. Two-sided 95% CIs are used for other efficacy endpoint estimates.

Results:

- 29 pts were enrolled from May 2016 to March 2023. During data validation and verification, 3 pts in this cohort were found to have tumors with *ERBB2/3* mutation (mut) only and were removed from this analysis. All 26 remaining pts were evaluable for efficacy.
- Pts had *ERBB2* amp only (n=17), *ERBB2* amp and mut (n=6), *ERBB2* amp and oe (n=1), *ERBB2* amp and *ERBB3* mut (n=1), and *ERBB3* amp only (n=1).
- **Demographics:** Median age 63 (range 29-88); 58% female; 89% self-identified as White, 8% as Black/African American, 4% preferred not to answer; 8% as Hispanic/Latino, 89% as not Hispanic/Latino, and 4% preferred not to answer.
- **Clinical Characteristics**: 77% PS 0-1, 23% PS 2; 50% received ≥3 prior systemic regimens. Primary tumor origin (# pts): cervix (6), esophagus (4), unspecified site (3), duodenum (2), urothelial carcinoma (2), vagina (2), lacrimal gland (1), ovary (1), peritoneum (1), small intestine (1), sweat gland (1), testes (1), and thymus (1).
- **Outcomes**: CR 2, PR 7, and SD16+ 7 for a DC rate of 62% (1-sided 90% CI, 47 to 100) (**Tables 1 and 2**). The null DC rate of ≤15% was rejected (p<0.001).
- Durations of CR were 48 and 151 wks, median duration of PR was 26 wks (range, 12-116), and median duration of SD for pts with SD16+ was 28 wks (range, 18-111).
- **Safety:** 5 pts (17%) had ≥1 drug-related grade 2-5 SAE or grade 3 AE including anemia, cardiac arrest resulting in death, diarrhea, ejection fraction decrease, pleural effusion, supraventricular tachycardia, and vomiting.

Conclusion: Pertuzumab plus trastuzumab demonstrated clinical activity in patients with various solid tumors with *ERBB2/3* amplification.

Table 1. Tumor Origin and Alterations of Pts Meeting Response Criteria (n=16)

Response	Primary Tumor Origin	Time to Progression, wks	*ERBB2/3* Alteration(s)	Co-alterations[a]
CR	Vagina	167	*ERBB2* amp, rearrangement[b]	*PIK3CA* amp
CR	Sweat gland	64	*ERBB2* amp	*TP53* N268fs, loss
PR	Esophagus	68	*ERBB2* amp	*TP53* V173L, D148fs*22
PR	Esophagus	40	*ERBB2* amp	*TP53* R196*
PR	Duodenum	28	*ERBB2* amp	*KRAS* G12D *TP53* Y205N
PR	Unspecified site	125	*ERBB2* amp, R678Q, V842I	*TP53* R342*
PR	Unspecified site	28	*ERBB2* amp	*HRAS* G13R, LOH *TP53* H193R, LOH
PR	Urothelial carcinoma	53	*ERBB2* amp *ERBB3* M120V	*ARID1A* S264* *TP53* E271Q, E285Q
PR	Vagina	42	*ERBB2* amp, oe	*PIK3R1* splice site 1745+2del, R461fs *TP53* R175H, oe
SD16+	Cervix	28	*ERBB2* amp	*TP53* Y220C
SD16+	Cervix	n/a[c]	*ERBB2* amp	--
SD16+	Duodenum	28	*ERBB3* amp	*KRAS* amp
SD16+	Small intestine	18	*ERBB2* amp	*TP53* E286fs*59
SD16+	Testes	24	*ERBB2* amp	--
SD16+	Unspecified site	28	*ERBB2* amp	*BRAF* F357Y[b] *TP53* R273C
SD16+	Urothelial carcinoma	18	*ERBB2* amp	*AKT2* E134*[b] *ARID1A* Q404* *PIK3CA* K111E

[a] Of the following genes examined: *AKT1, AKT2, AKT3, ALK, ARID1A, BRAF, EGFR, HRAS, KRAS, MET, NRAS, PIK3CA, PIK3R1, PTEN, TP53*
[b] Variant of unknown significance
[c] 113 wks as of April 22nd, 2024

Funding supported by Genentech. The authors would like to thank the patients who participated in this cohort, the clinical centers and staff, as well as Tania Szado, PhD, clinical lead of Genentech, a TAPUR participating pharmaceutical company.
Contact: TAPURPublications@asco.org

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® and the author of this poster.

Table 2: Efficacy Outcomes (N=26)

DC rate, % (1-sided 90% CI) (p-value)	62 (47, 100) (p<0.001)
OR rate, % (95% CI)	31 (17, 56)
Median PFS, wks (95% CI)	26 (12, 28)
Median OS, wks (95% CI)	64 (34, 140)
Duration of CR, wks (n=2)	48 and 151
Median duration of PR (range), wks (n=7)	26 (12, 116)
Median duration of OR (range), wks (n=9)	45 (12, 159)
Median duration of SD in pts with SD16+, wks (n=7)	28 (18, 111)

Figure 1: Best Percent Change from Baseline in Target Lesion Size (N=26)

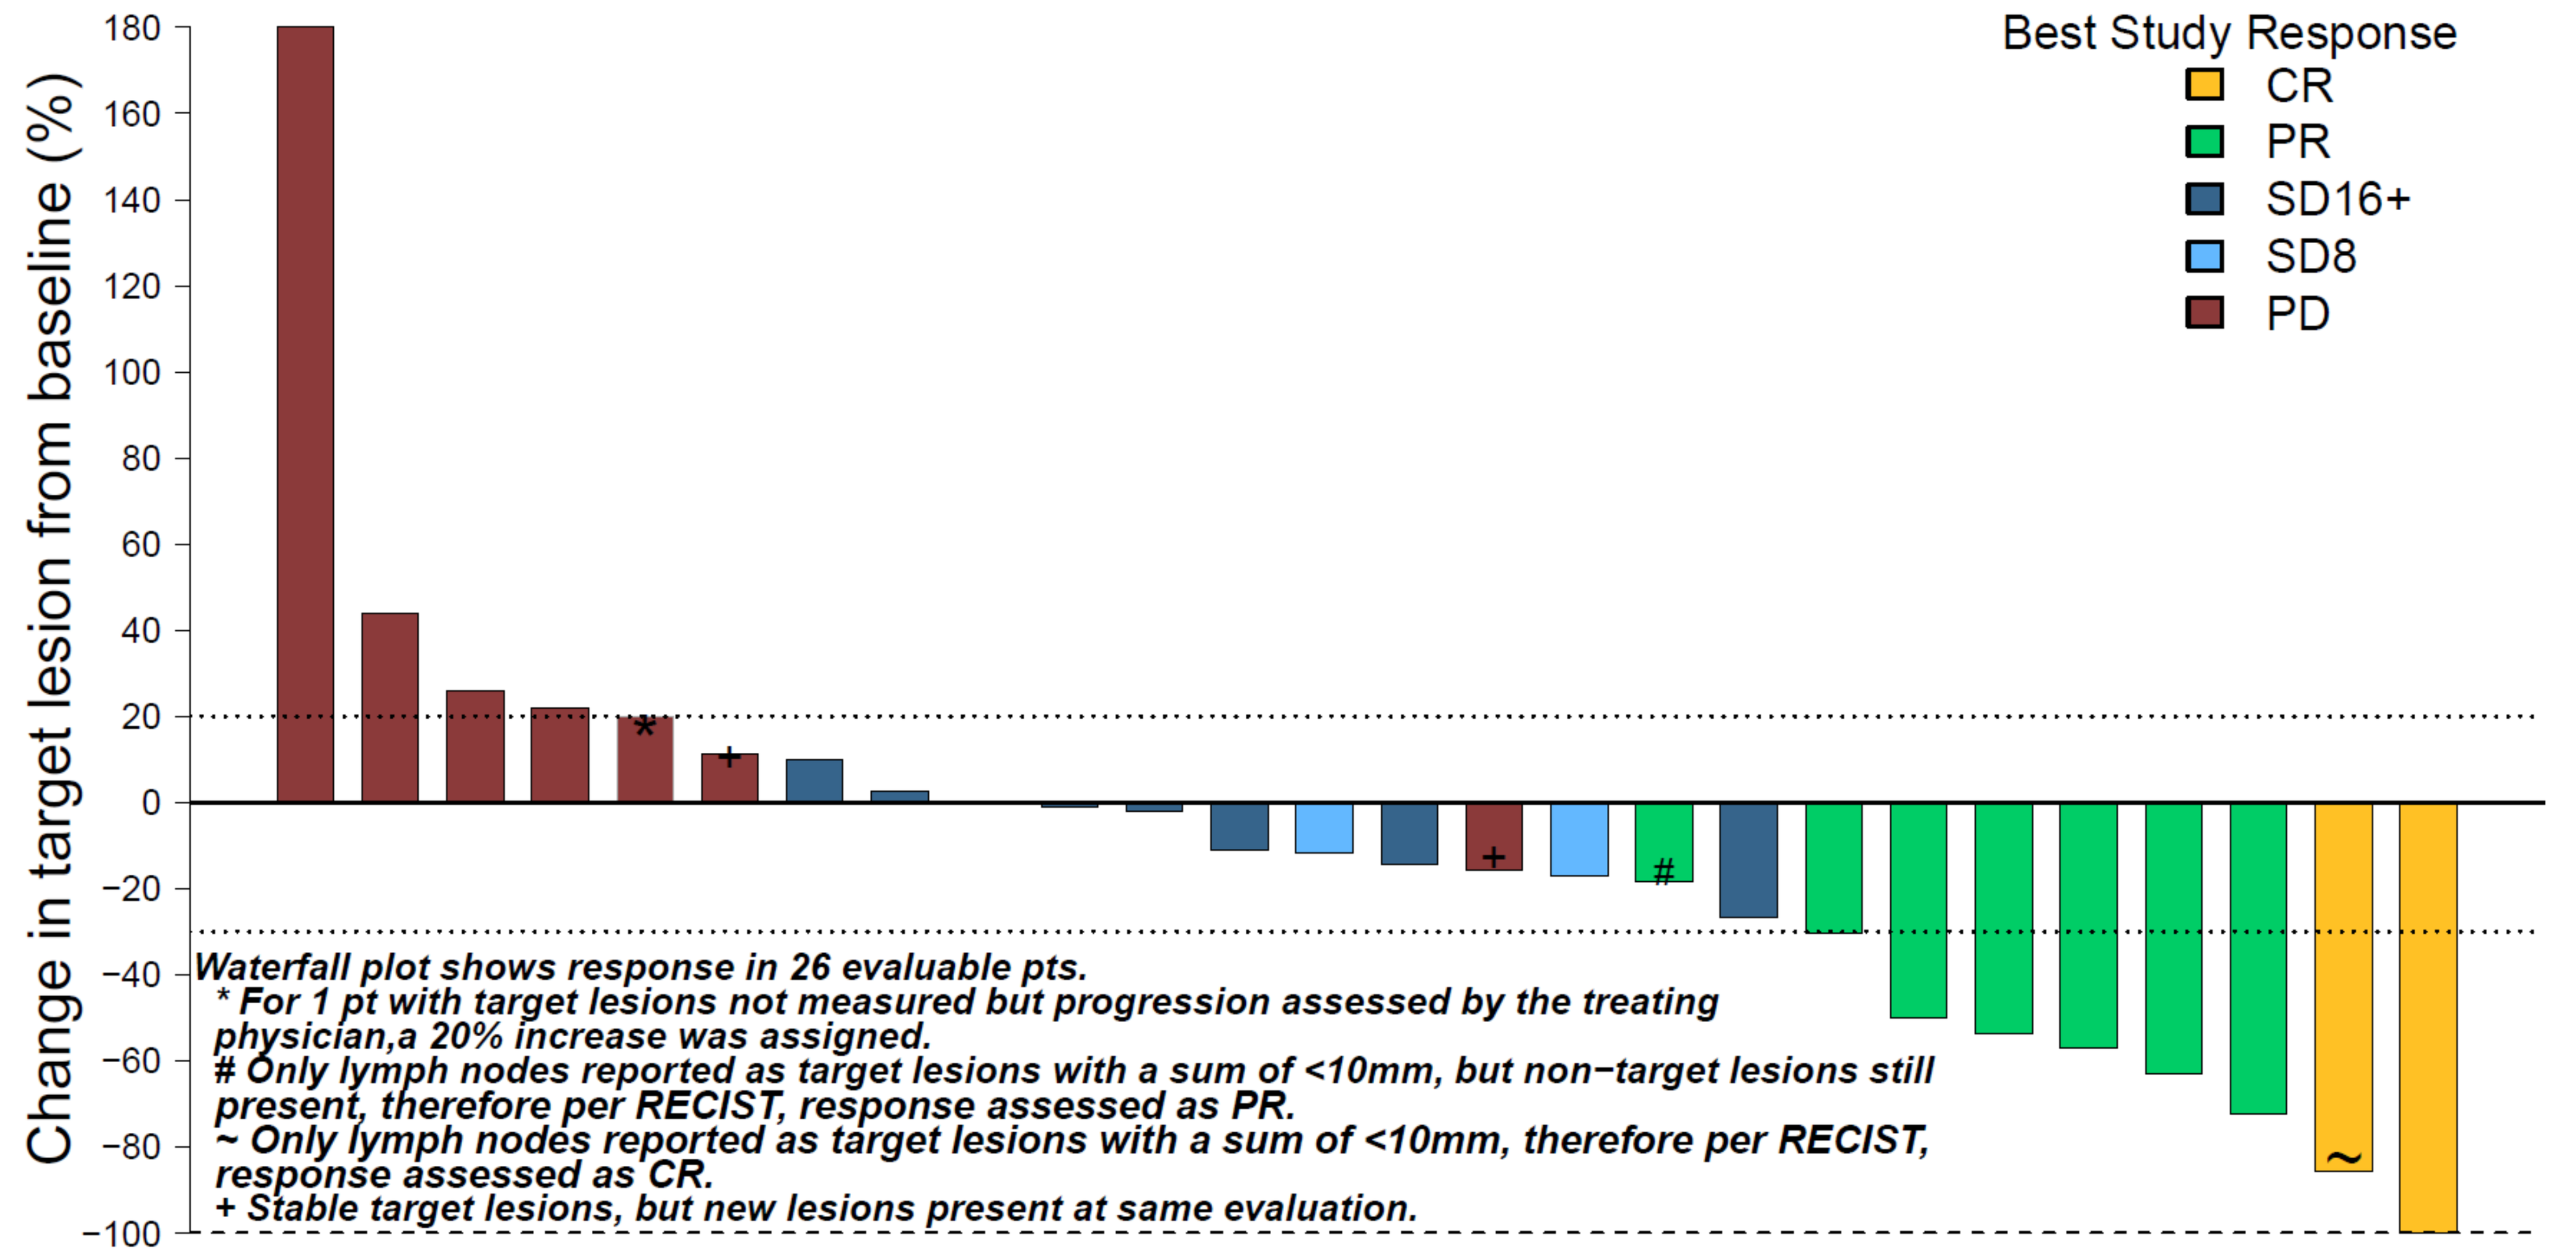
Change in target lesion from baseline (%)
180
160
140
120
100
80
60
40
20
0
−20
−40
−60
−80
−100
Best Study Response
CR
PR
SD16+
SD8
PD
*
+
+
#
~
Waterfall plot shows response in 26 evaluable pts.
* For 1 pt with target lesions not measured but progression assessed by the treating physician,a 20% increase was assigned.
Only lymph nodes reported as target lesions with a sum of <10mm, but non−target lesions still present, therefore per RECIST, response assessed as PR.
~ Only lymph nodes reported as target lesions with a sum of <10mm, therefore per RECIST, response assessed as CR.
+ Stable target lesions, but new lesions present at same evaluation.

Figure 2: Time on Treatment for Pts with OR or SD16+ (n=16)

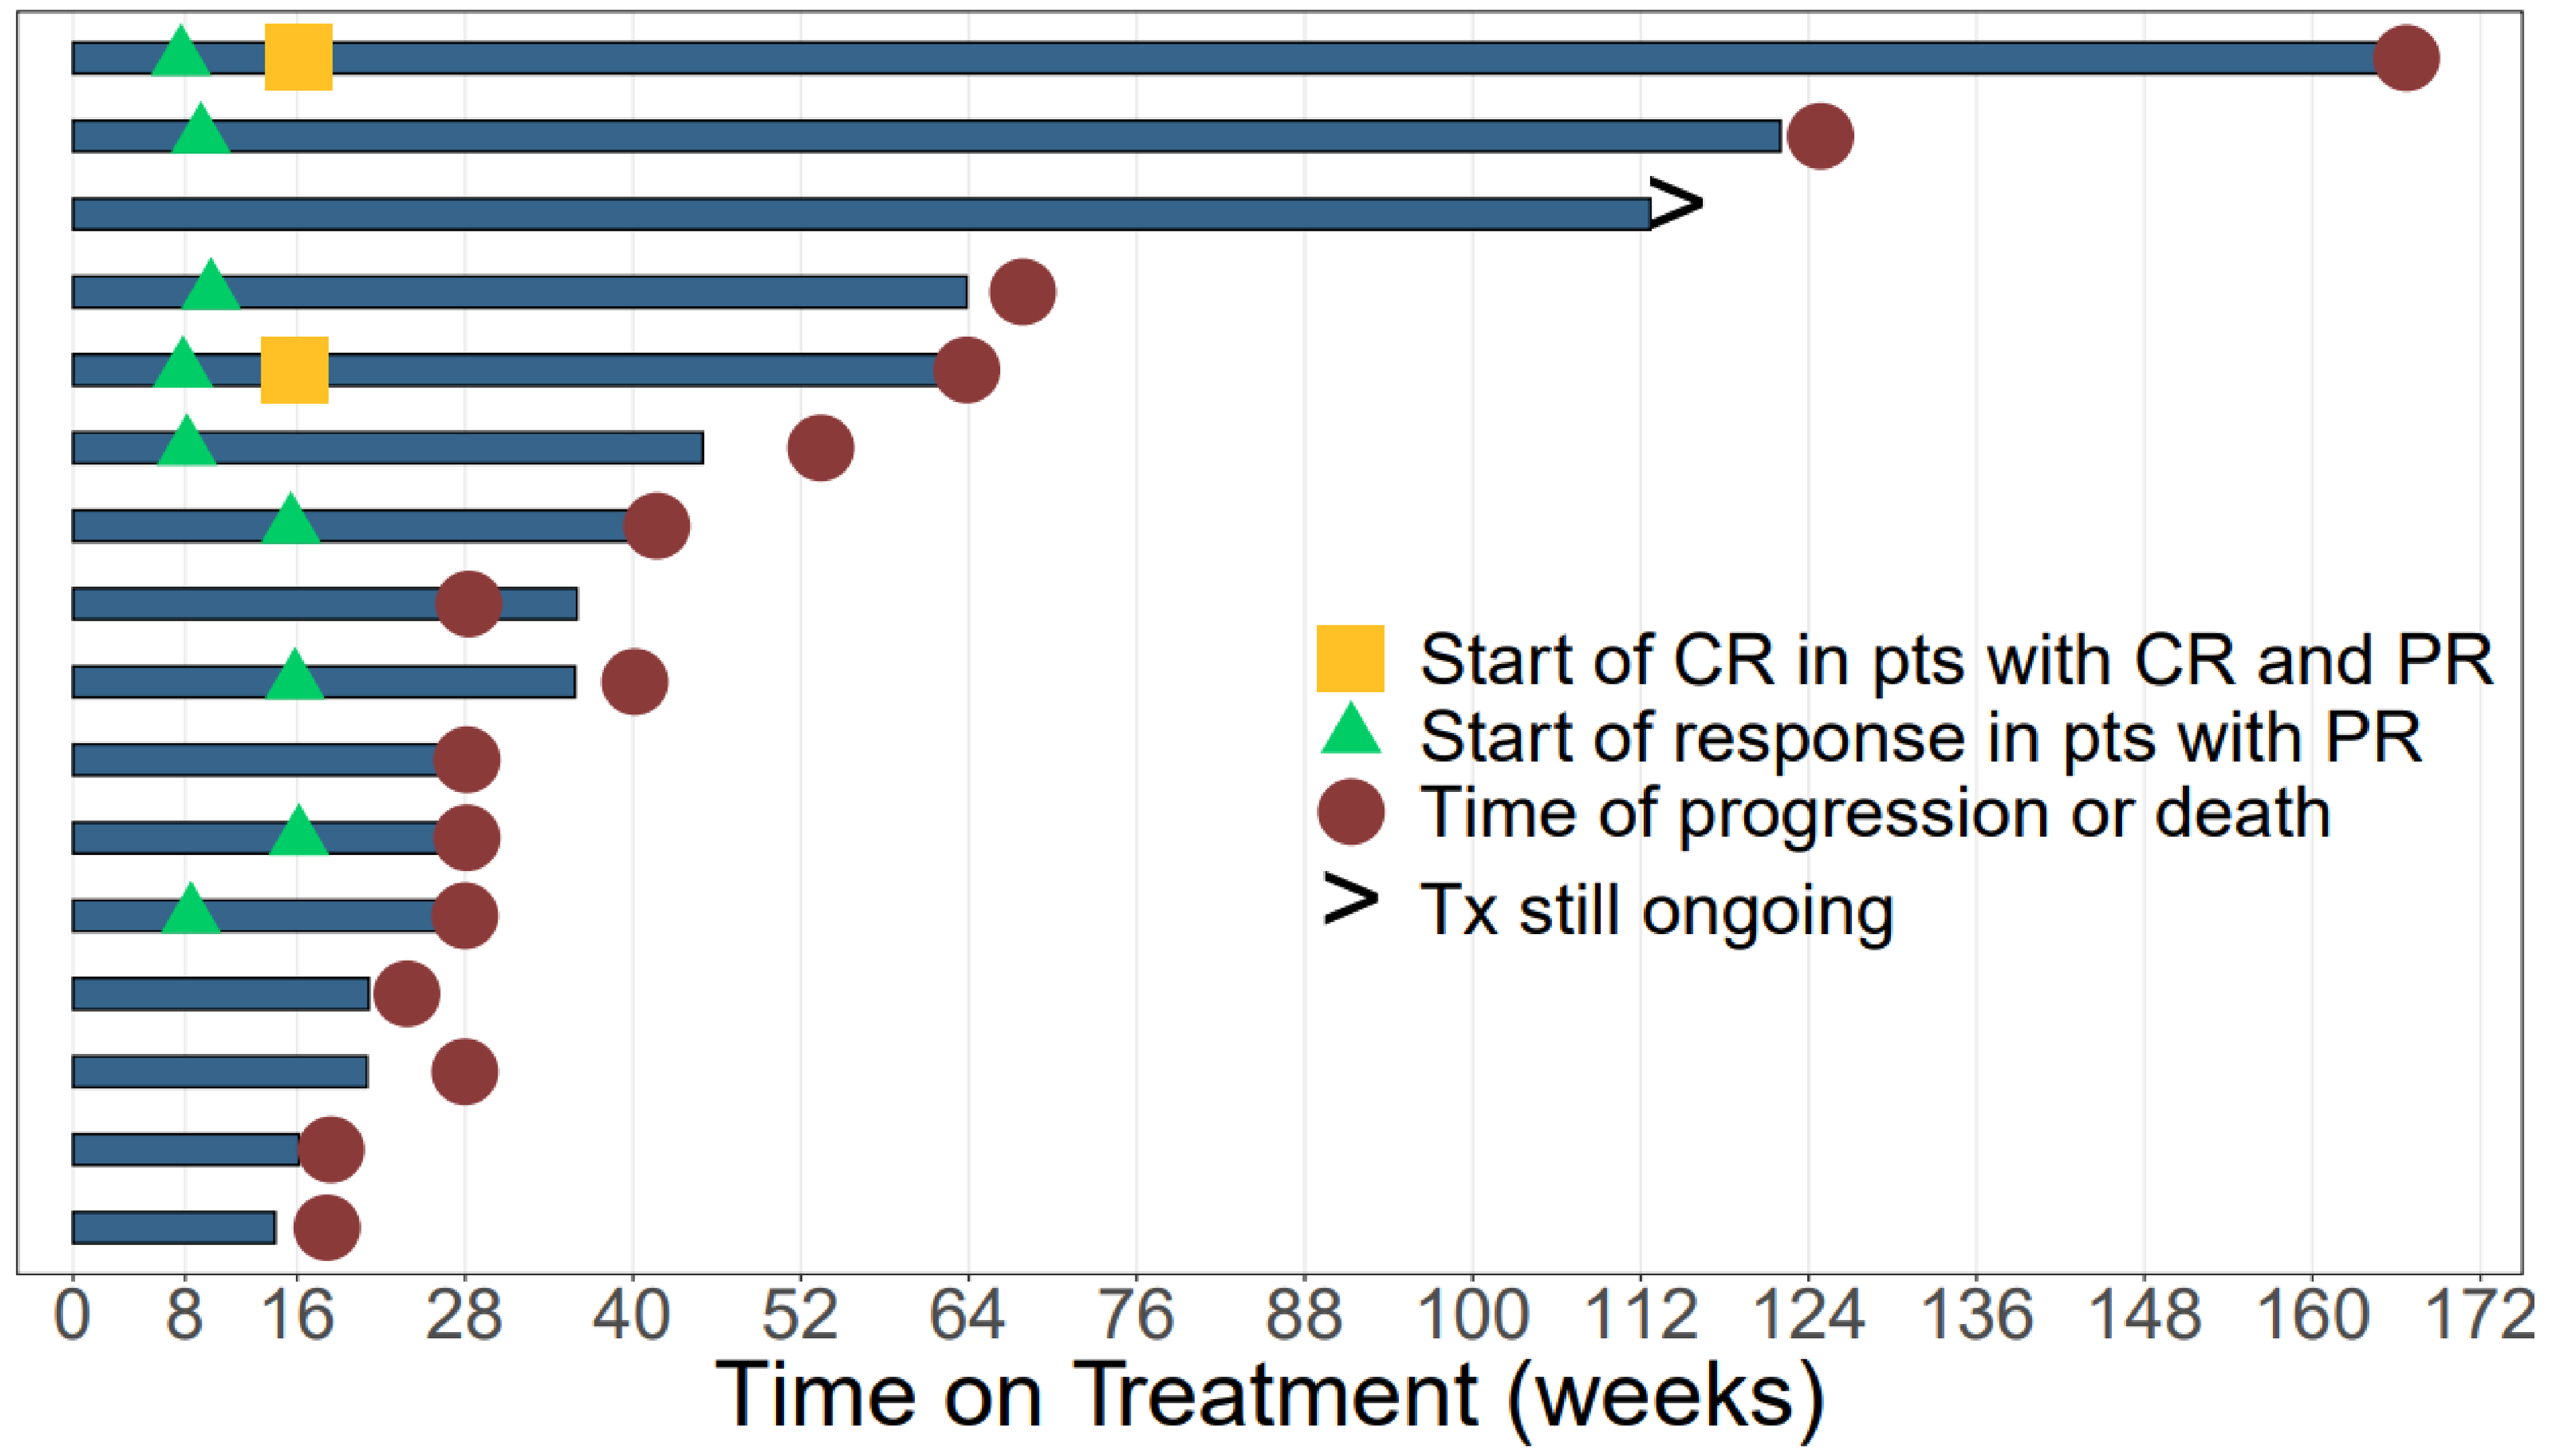
Start of CR in pts with CR and PR
Start of response in pts with PR
Time of progression or death
> Tx still ongoing
0
8
16
28
40
52
64
76
88
100
112
124
136
148
160
172
Time on Treatment (weeks)